POSTER NUMBER 7

Plecanatide Is Efficacious in Patients With Irritable Bowel Syndrome With Constipation and Bloating: Evaluation Using Trisymptom Composite Endpoints

Darren M. Brenner, MD[1]; Andrea S. Shin, MD[2]; Adam P. Laitman, MD[3]; Stacey S. Hodges, DNP[3]; David C. Kunkel, MD[4]

[1]Northwestern University, Chicago, IL; [2]University of California Los Angeles, Los Angeles, CA; [3]Salix Pharmaceuticals, Bridgewater, NJ; [4]University of California San Diego, La Jolla, CA

INTRODUCTION

- Patients with irritable bowel syndrome (IBS) commonly have sensory-related symptoms (eg, abdominal pain, bloating)[1]
- Along with abdominal pain, bloating is one of the most common symptoms identified in patients with IBS with constipation (IBS-C), with most reporting moderate to severe bloating[2]
- Abdominal pain/discomfort and bloating prompt many patients to seek medical care[3,4]: it is important to identify medications that effectively address and improve the cardinal bowel and abdominal symptoms of IBS
- Plecanatide (Trulance®, Salix Pharmaceuticals, Bridgewater, NJ), a guanylate cyclase-C agonist, is indicated for the treatment of chronic idiopathic constipation and IBS-C in adults (3 mg once daily)[5]
 - Phase 3, randomized, placebo-controlled trials have demonstrated that plecanatide improves multiple objective gastrointestinal symptoms, including bowel movement frequency[6-8]
 - In addition, plecanatide has been shown to improve IBS-C symptoms in patients across varying levels of baseline bloating severity[9]

AIM

- To evaluate the efficacy of plecanatide in an IBS-C population with bloating utilizing a novel trisymptom composite endpoint (ie, abdominal pain, bloating, and complete spontaneous bowel movements [CSBMs]) at various thresholds of response

METHODS

- Data were pooled and analyzed post hoc from 2 identically designed, phase 3, randomized, placebo-controlled trials[6]
- Population included in the current analyses were males or females aged 18 to 40 years with IBS-C (Rome III), baseline bloating (score ≥1 on a numeric rating scale ranging from 0 ["No"] to 10 ["Worst Possible"]), and body mass index between 18 and 40 kg/m^2
 - Patients were treated with plecanatide 3 mg or placebo once daily for 12 weeks*
- Abdominal pain and bloating intensity (11-point scale, range, 0 ["no"] to 10 ["worst possible"] for each) and frequency and completeness of bowel movements were recorded in a daily diary

*Patients in the plecanatide 6-mg treatment arm of the original trials were not included in the current analyses.[6]

- Response was defined as simultaneous improvement from baseline in all 3 symptoms (abdominal pain, bloating, and CSBMs/wk) for ≥6 of the 12 weeks of treatment
 - Several composite criteria thresholds were evaluated (ie, ≥2-point or ≥30% or ≥40% improvement in abdominal pain and bloating plus an increase of ≥1 or ≥2 CSBMs in the same week for ≥6 of 12 weeks)
- Patients were also stratified by baseline bloating intensity (mild [score, 1-5]; moderate/severe [score, 6-10])
- *P* values were calculated using the Cochran-Mantel-Haenszel test, with stratification by gender

RESULTS

- 605 patients (aged 18-40 years) with IBS-C and baseline bloating were included in the analysis (Table)
 - The majority (71.7%) of the 605 patients had moderate to severe bloating

Table. Demographics and Baseline Characteristics

Parameter	Plecanatide 3 mg (n=313)	Placebo (n=292)
Age, y		
Mean (SD)	30.6 (6.3)	30.3 (6.6)
Median (range)	31.0 (18-40)	31.0 (18-40)
Female, n (%)	220 (70.3)	213 (72.9)
BMI, kg/m^2, mean (SD)	27.3 (4.8)	27.0 (4.6)
Race/ethnicity, n (%)		
White	237 (75.7)	226 (77.4)
Black	59 (18.8)	54 (18.5)
Asian	13 (4.2)	8 (2.7)
Other	4 (1.3)	4 (1.4)
Abdominal pain score, mean (SD)*	6.2 (1.7)	6.4 (1.7)
Bloating score*		
Mean (SD)	6.4 (1.7)	6.6 (1.7)
Mild intensity—score 1-5, n (%)	94 (30.0)	77 (26.4)
Moderate/severe intensity—score 6-10, n (%)	219 (70.0)	215 (73.6)
CSBMs/week, mean (SD)*	0.2 (0.5)	0.2 (0.5)

*Abdominal pain and bloating were measured using an 11-point scale (range, 0 ["no"] to 10 ["worst possible"]).
BMI = body mass index; CSBM = complete spontaneous bowel movement.

RESULTS

- Overall, a significantly greater percentage of patients treated with plecanatide compared with placebo were trisymptom composite responders, defined using several stringent thresholds (Figure 1)

Figure 1. IBS-C Trisymptom Composite (Abdominal Pain, Bloating, and CSBM) Responders (Overall Population)

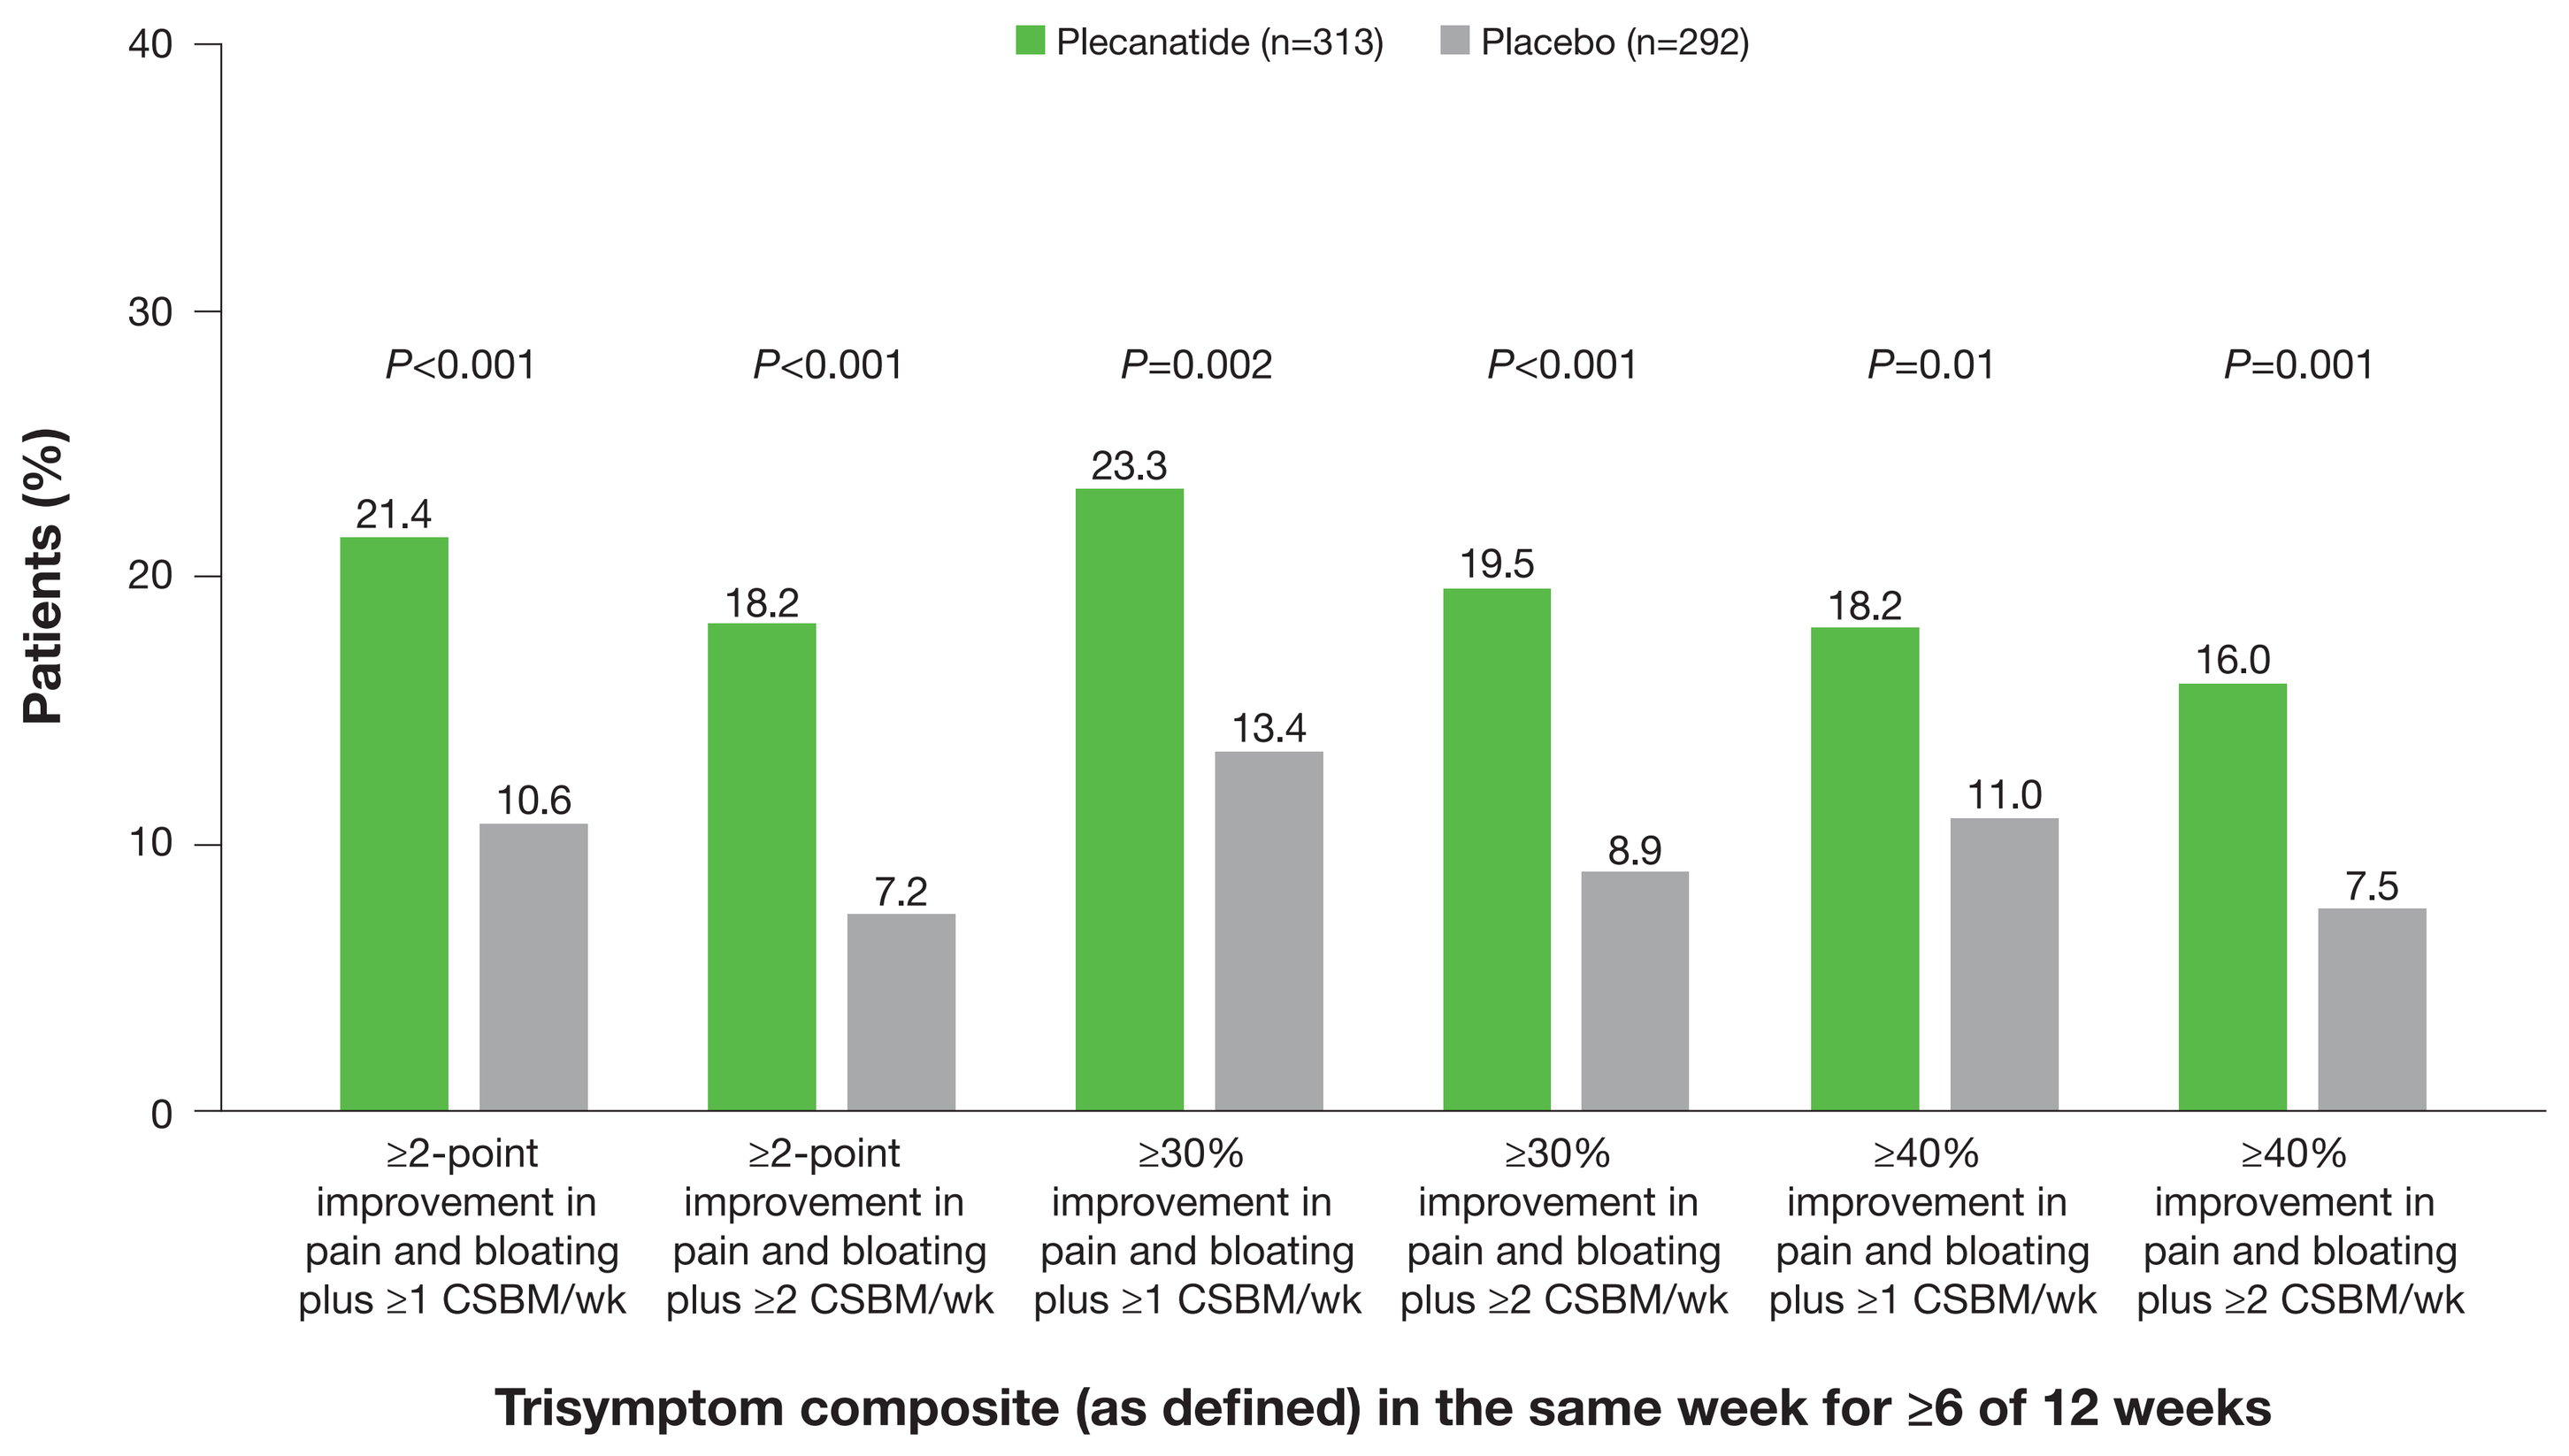
Plecanatide (n=313)
Placebo (n=292)
Patients (%)
0
10
20
30
40
P<0.001
P<0.001
P=0.002
P<0.001
P=0.01
P=0.001
21.4
10.6
18.2
7.2
23.3
13.4
19.5
8.9
18.2
11.0
16.0
7.5
≥2-point improvement in pain and bloating plus ≥1 CSBM/wk
≥2-point improvement in pain and bloating plus ≥2 CSBM/wk
≥30% improvement in pain and bloating plus ≥1 CSBM/wk
≥30% improvement in pain and bloating plus ≥2 CSBM/wk
≥40% improvement in pain and bloating plus ≥1 CSBM/wk
≥40% improvement in pain and bloating plus ≥2 CSBM/wk
Trisymptom composite (as defined) in the same week for ≥6 of 12 weeks

CSBM = complete spontaneous bowel movement; IBS-C = irritable bowel syndrome with constipation.

- When data were stratified by baseline bloating intensity (mild; moderate/severe), significant improvements favoring plecanatide compared with placebo were noted for most of the stringent trisymptom composite endpoints analyzed (Figure 2A and 2B)

Figure 2. IBS-C Trisymptom Composite Responders (Abdominal Pain, Bloating, and CSBM) in Patients With Mild (A) or Moderate/Severe (B) Baseline Bloating Intensity

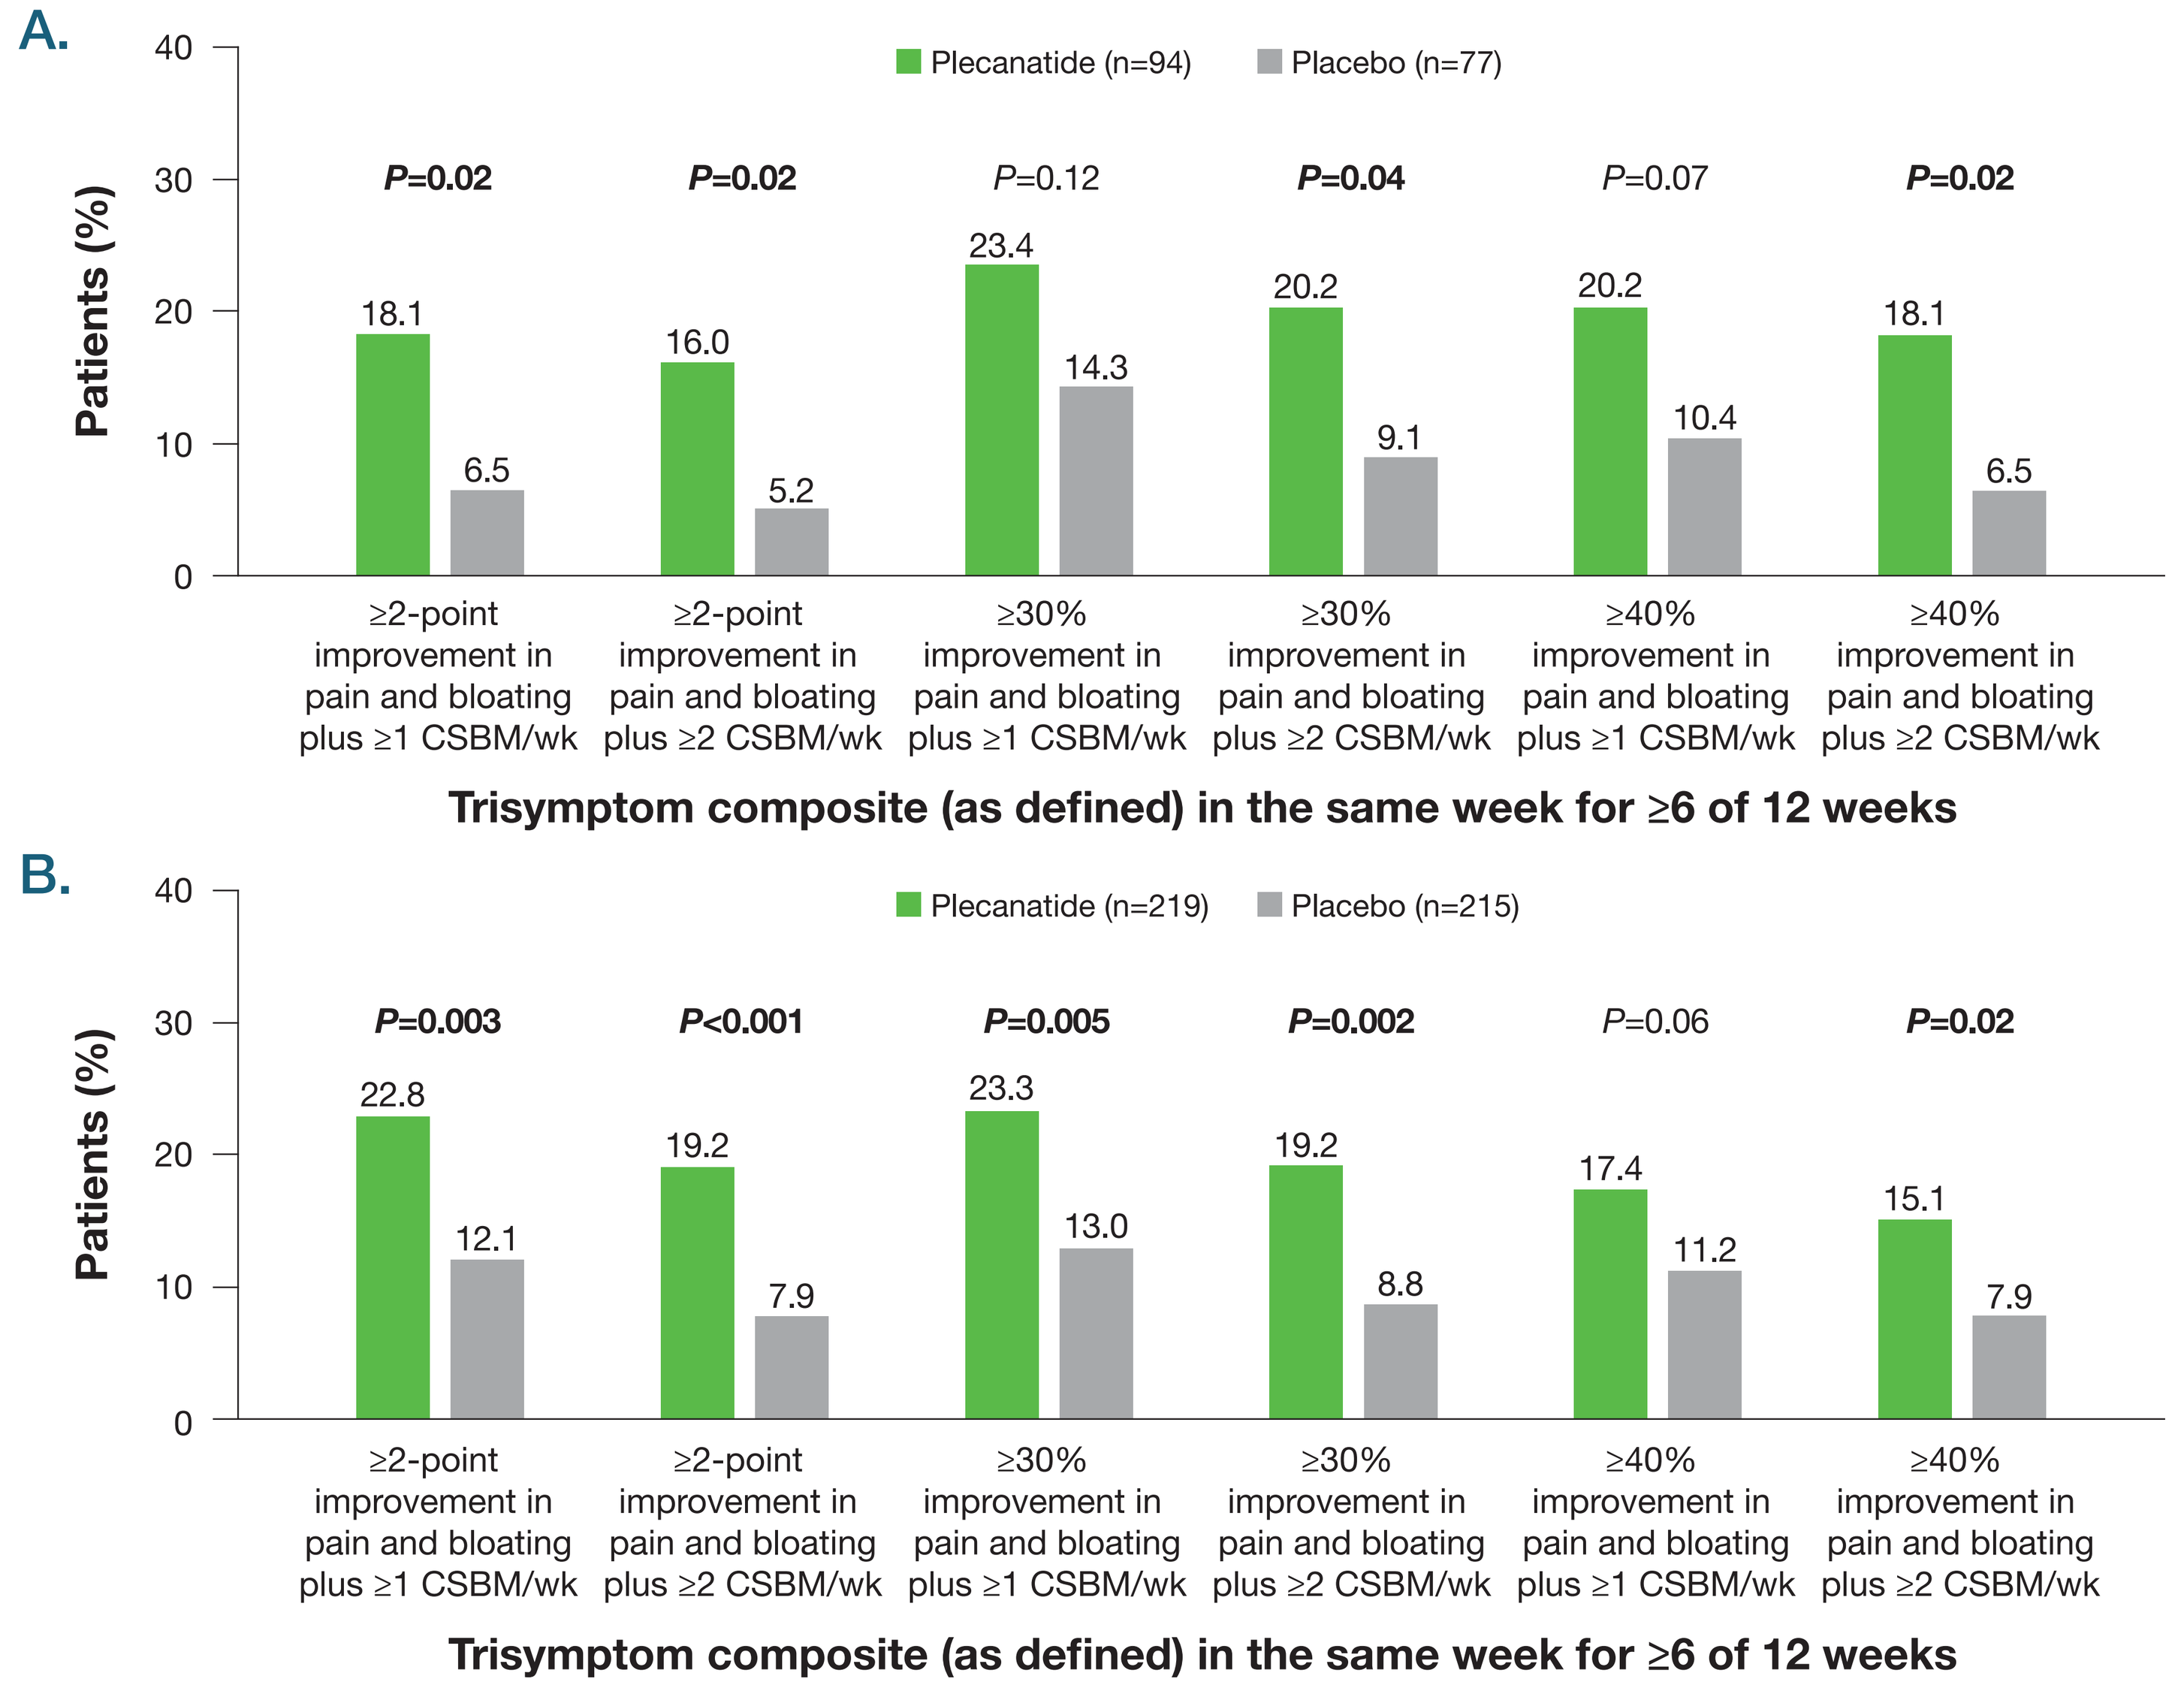
A.
Plecanatide (n=94)
Placebo (n=77)
Patients (%)
0
10
20
30
40
P=0.02
P=0.02
P=0.12
P=0.04
P=0.07
P=0.02
18.1
6.5
16.0
5.2
23.4
14.3
20.2
9.1
20.2
10.4
18.1
6.5
≥2-point improvement in pain and bloating plus ≥1 CSBM/wk
≥2-point improvement in pain and bloating plus ≥2 CSBM/wk
≥30% improvement in pain and bloating plus ≥1 CSBM/wk
≥30% improvement in pain and bloating plus ≥2 CSBM/wk
≥40% improvement in pain and bloating plus ≥1 CSBM/wk
≥40% improvement in pain and bloating plus ≥2 CSBM/wk
Trisymptom composite (as defined) in the same week for ≥6 of 12 weeks
B.
Plecanatide (n=219)
Placebo (n=215)
Patients (%)
0
10
20
30
40
P=0.003
P<0.001
P=0.005
P=0.002
P=0.06
P=0.02
22.8
12.1
19.2
7.9
23.3
13.0
19.2
8.8
17.4
11.2
15.1
7.9
≥2-point improvement in pain and bloating plus ≥1 CSBM/wk
≥2-point improvement in pain and bloating plus ≥2 CSBM/wk
≥30% improvement in pain and bloating plus ≥1 CSBM/wk
≥30% improvement in pain and bloating plus ≥2 CSBM/wk
≥40% improvement in pain and bloating plus ≥1 CSBM/wk
≥40% improvement in pain and bloating plus ≥2 CSBM/wk
Trisymptom composite (as defined) in the same week for ≥6 of 12 weeks

Bold *P* values indicate statistical significance.
CSBM = complete spontaneous bowel movement; IBS-C = irritable bowel syndrome with constipation.

CONCLUSIONS

- Plecanatide simultaneously and significantly improved abdominal pain, bloating, and CSBM frequency, using varying thresholds to define trisymptom composite responses
 - These responses were maintained regardless of baseline bloating intensity
- Plecanatide appears effective for treating global and individual sensatory and bowel symptoms characteristic of IBS in patients with IBS-C who reported any bloating at the start of therapy

REFERENCES: **1.** Lacy BE, Mearin F, Chang L, et al. *Gastroenterology.* 2016;150(6):1393-1407. **2.** Bhinder G, Meza-Cardona JM, Low A, et al. *J Can Assoc Gastroenterol.* 2023;6(6):219-228. **3.** Jiang X, Locke GR, Zinsmeister AR, et al. *Aliment Pharmacol Ther.* 2009;30(7):775-783. **4.** Ringel Y, Williams RE, Kalilani L, et al. *Clin Gastroenterol Hepatol.* 2009;7(1):68-72. **5.** Trulance (plecanatide) tablets, for oral use [package insert]. Bridgewater, NJ: Salix Pharmaceuticals; 2024. **6.** Brenner DM, Fogel R, Dorn SD, et al. *Am J Gastroenterol.* 2018;113(5):735-745. **7.** Miner PB Jr, Koltun WD, Wiener GJ, et al. *Am J Gastroenterol.* 2017;112(4):613-621. **8.** DeMicco M, Barrow L, Hickey B, et al. *Therap Adv Gastroenterol.* 2017;10(11):837-851. **9.** Brenner DM, Sharma A, Rao SSC, et al. *Dig Dis Sci.* 2024;69(5):1731-1738.

ACKNOWLEDGMENTS: The post hoc analyses were supported by Salix Pharmaceuticals. Technical editorial and medical writing assistance were provided under direction of the authors by Mary Beth Moncrief, PhD, Synchrony Medical Communications, LLC, West Chester, PA. Funding for this assistance was provided by Salix Pharmaceuticals.

DISCLOSURES: DMB reports being a consultant and speaker for Salix Pharmaceuticals. ASS reports serving on an Ardelyx Scientific Communications Advisory Board for irritable bowel syndrome with constipation. APL and SSH are employees of Salix Pharmaceuticals. DCK reports being a consultant for Ardelyx, Evoke Pharma, Mahana Therapeutics, Inc., and Pfizer Inc., and serving as a speaker for Regeneron Pharmaceuticals Inc.

American Association of Nurse Practitioners (AANP) National Conference • June 17–22, 2025 • San Diego, CA

Research funded by: Salix Pharmaceuticals